Abstract #LB071

Recursion

A Phenomics Platform Combining Imaging and Artificial Intelligence for Rapid Validation and Advancement of Novel Oncology Targets

Jenny Rudnick[1], Kiran Nadella[1], Malini Rajan[1], Chase Neumann[1], Shane Rowley[1], Ethan Gardner[1], Shadi Swaidani[1], Aimee Iberg[1], Lu Chen[1], Daria Beshnova[1], Aurora Blucher[1], Rebecca Sarto Basso[1], Kevin Fales[1], Ashraf Saeed[1], Christopher Bailey[1], Weston Judd[1], Chrissy Egbert[1], Joel Ellis[1], John Ansede[1], Pouya Hadipour[1], Kevin Jessing[1], Janet Paulsen[1], Paul Rearden[1], Vamshi Manda[1], Sashi Kasimshetty[1], Michael Hancock[1], Harish Shankaran[1], Bryan Ellis[1], Meenakshy Iyer[1], Carl Brooks[1], Ashish Bhandari[1], Chris Gibson[1], Irit Rappley[1], Joe Carpenter[1], Laura Schaevitz[1], Imran Haque[1], Hayley Donnella[1], Michael Cuccarese[1], Marie Evangelista[1]

[1]Recursion Pharmaceuticals, Inc. Salt Lake City, UT.

0 ABSTRACT

The emergence of technological innovations has created the opportunity to envision new approaches to discover therapeutics at scale. We combined advances in high content microscopy with arrayed CRISPR genome editing techniques and machine learning (ML) to build a rigorously controlled dataset enabling exploration of biology and chemistry at scale. Phenotypes from millions of perturbations in multiple cell types were embedded in a unified representation space and leveraged to accelerate discovery and reverse translation, ultimately yielding novel biological insights, and optimizing the advancement of lead molecular series through structure-activity relationships (SAR). Here, we demonstrate the capability of our platform to discover potential cancer therapies with distinct mechanisms of action. First, we describe the identification of a novel compound series that potentiates the effects of immunotherapy in syngeneic mouse models, producing complete responses and immunological memory, while also limiting peripheral inflammation. Specific novel chemical entities (NCEs) caused robust CD45+ cell influx into the tumor microenvironment and significantly attenuated exhausted T cells and immunosuppressive macrophages, thereby enhancing anti-tumor immunity. Strikingly, the same NCEs suppressed peripheral inflammation while sustaining elevated levels of intra-tumoral proinflammatory cytokines. Second, we highlight a novel and differentiated strategy to potentiate PARP inhibitor response in homologous repair deficient (HRD) - negative or HR-proficient ovarian cancers. NCEs altered the expression of genes within the DNA damage repair (DDR) network and cell cycle checkpoints to synergize with PARP inhibition in vivo and resensitized a PARP-resistant patient-derived xenograft (PDX) model. Collectively, we believe future efforts on the industrialization and integration of various technological innovations across biology, chemistry, automation, data science, and engineering will ultimately modernize drug discovery and radically improve patient lives.

1 RECURSION'S SCREENING PLATFORM CAPTURES A HIGH DIMENSIONAL PHENOTYPIC READOUT FROM HUMAN CELLS AT MASSIVE SCALE, REFLECTING KNOWN AND NOVEL BIOLOGICAL RELATIONSHIPS

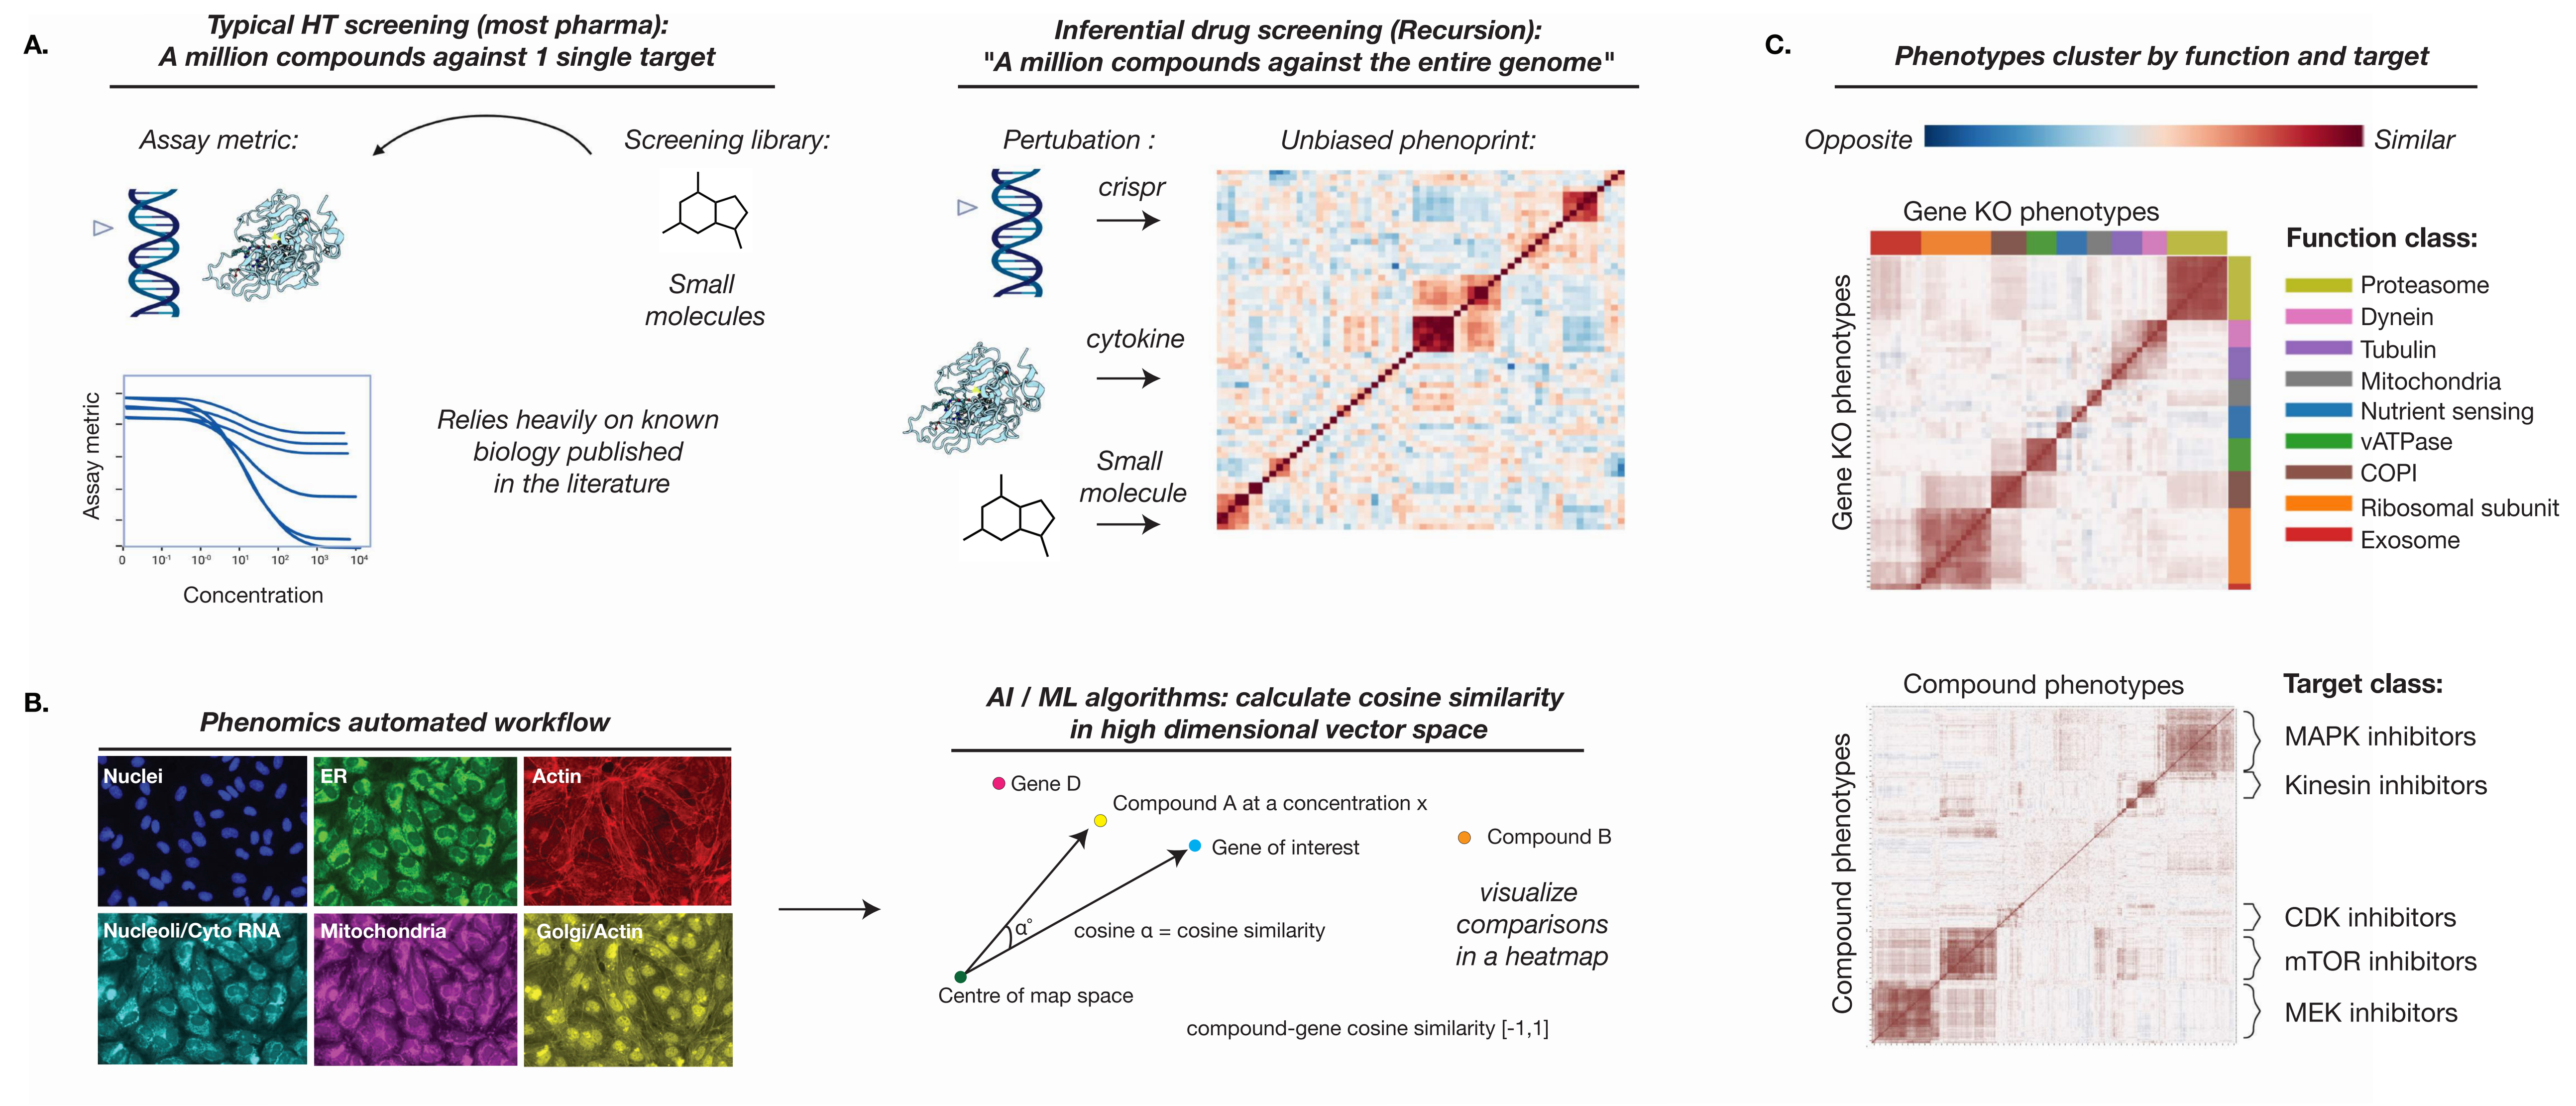

A.
Typical HT screening (most pharma): A million compounds against 1 single target
Assay metric:
Screening library:
Small molecules
Assay metric
Concentration
Relies heavily on known biology published in the literature
Inferential drug screening (Recursion): "A million compounds against the entire genome"
Pertubation :
Unbiased phenoprint:
crispr
cytokine
Small molecule
B.
Phenomics automated workflow
Nuclei
ER
Actin
Nucleoli/Cyto RNA
Mitochondria
Golgi/Actin
AI / ML algorithms: calculate cosine similarity in high dimensional vector space
Gene D
Compound A at a concentration x
Gene of interest
Compound B
cosine α = cosine similarity
Centre of map space
visualize comparisons in a heatmap
compound-gene cosine similarity [-1,1]
C.
Phenotypes cluster by function and target
Opposite
Similar
Gene KO phenotypes
Function class:
Proteasome
Dynein
Tubulin
Mitochondria
Nutrient sensing
vATPase
COPI
Ribosomal subunit
Exosome
Compound phenotypes
Target class:
MAPK inhibitors
Kinesin inhibitors
CDK inhibitors
mTOR inhibitors
MEK inhibitors

Figure 1: An overview of Recursion's phenomic platform for scaled discovery and exploration. (A) The traditional pharma approach to drug discovery typically involves a biased "one target approach," utilizing a small molecule library to modulate one target with a specific assay readout, such as a change in gene expression, cell viability or proliferation. This approach is time consuming and expensive. Recursion's approach to drug discovery utilizes "inferential drug screening." We treat various human cell types with a range of biological perturbants such as Crispr-based knockouts, cytokines or small molecules to generate high throughput fluorescence microscopy deep-learning enabled images called "phenoprints." (B) Vector representations of millions of multi-channel fluorescence microscopy images generated using a proprietary analytics workflow allow us to generate heat maps of gene-gene and gene-compound relationships. AI/ML algorithms are used to calculate cosine similarity in high dimensional vector space. (C) Top, gene knockouts with similar cellular phenotypes yield similar phenoprints. For example, genes that encode for proteosome associated proteins form a distinct and strong phenosimilar cluster (yellow). Bottom, clustering of compound phenotypes captures known mechanism of action and groups compounds together based on shared mechanism.

2 USING BIOLOGICAL MAPS TO IDENTIFY NOVEL TARGETS AND DRIVE ITERATIVE CHEMICAL OPTIMIZATION

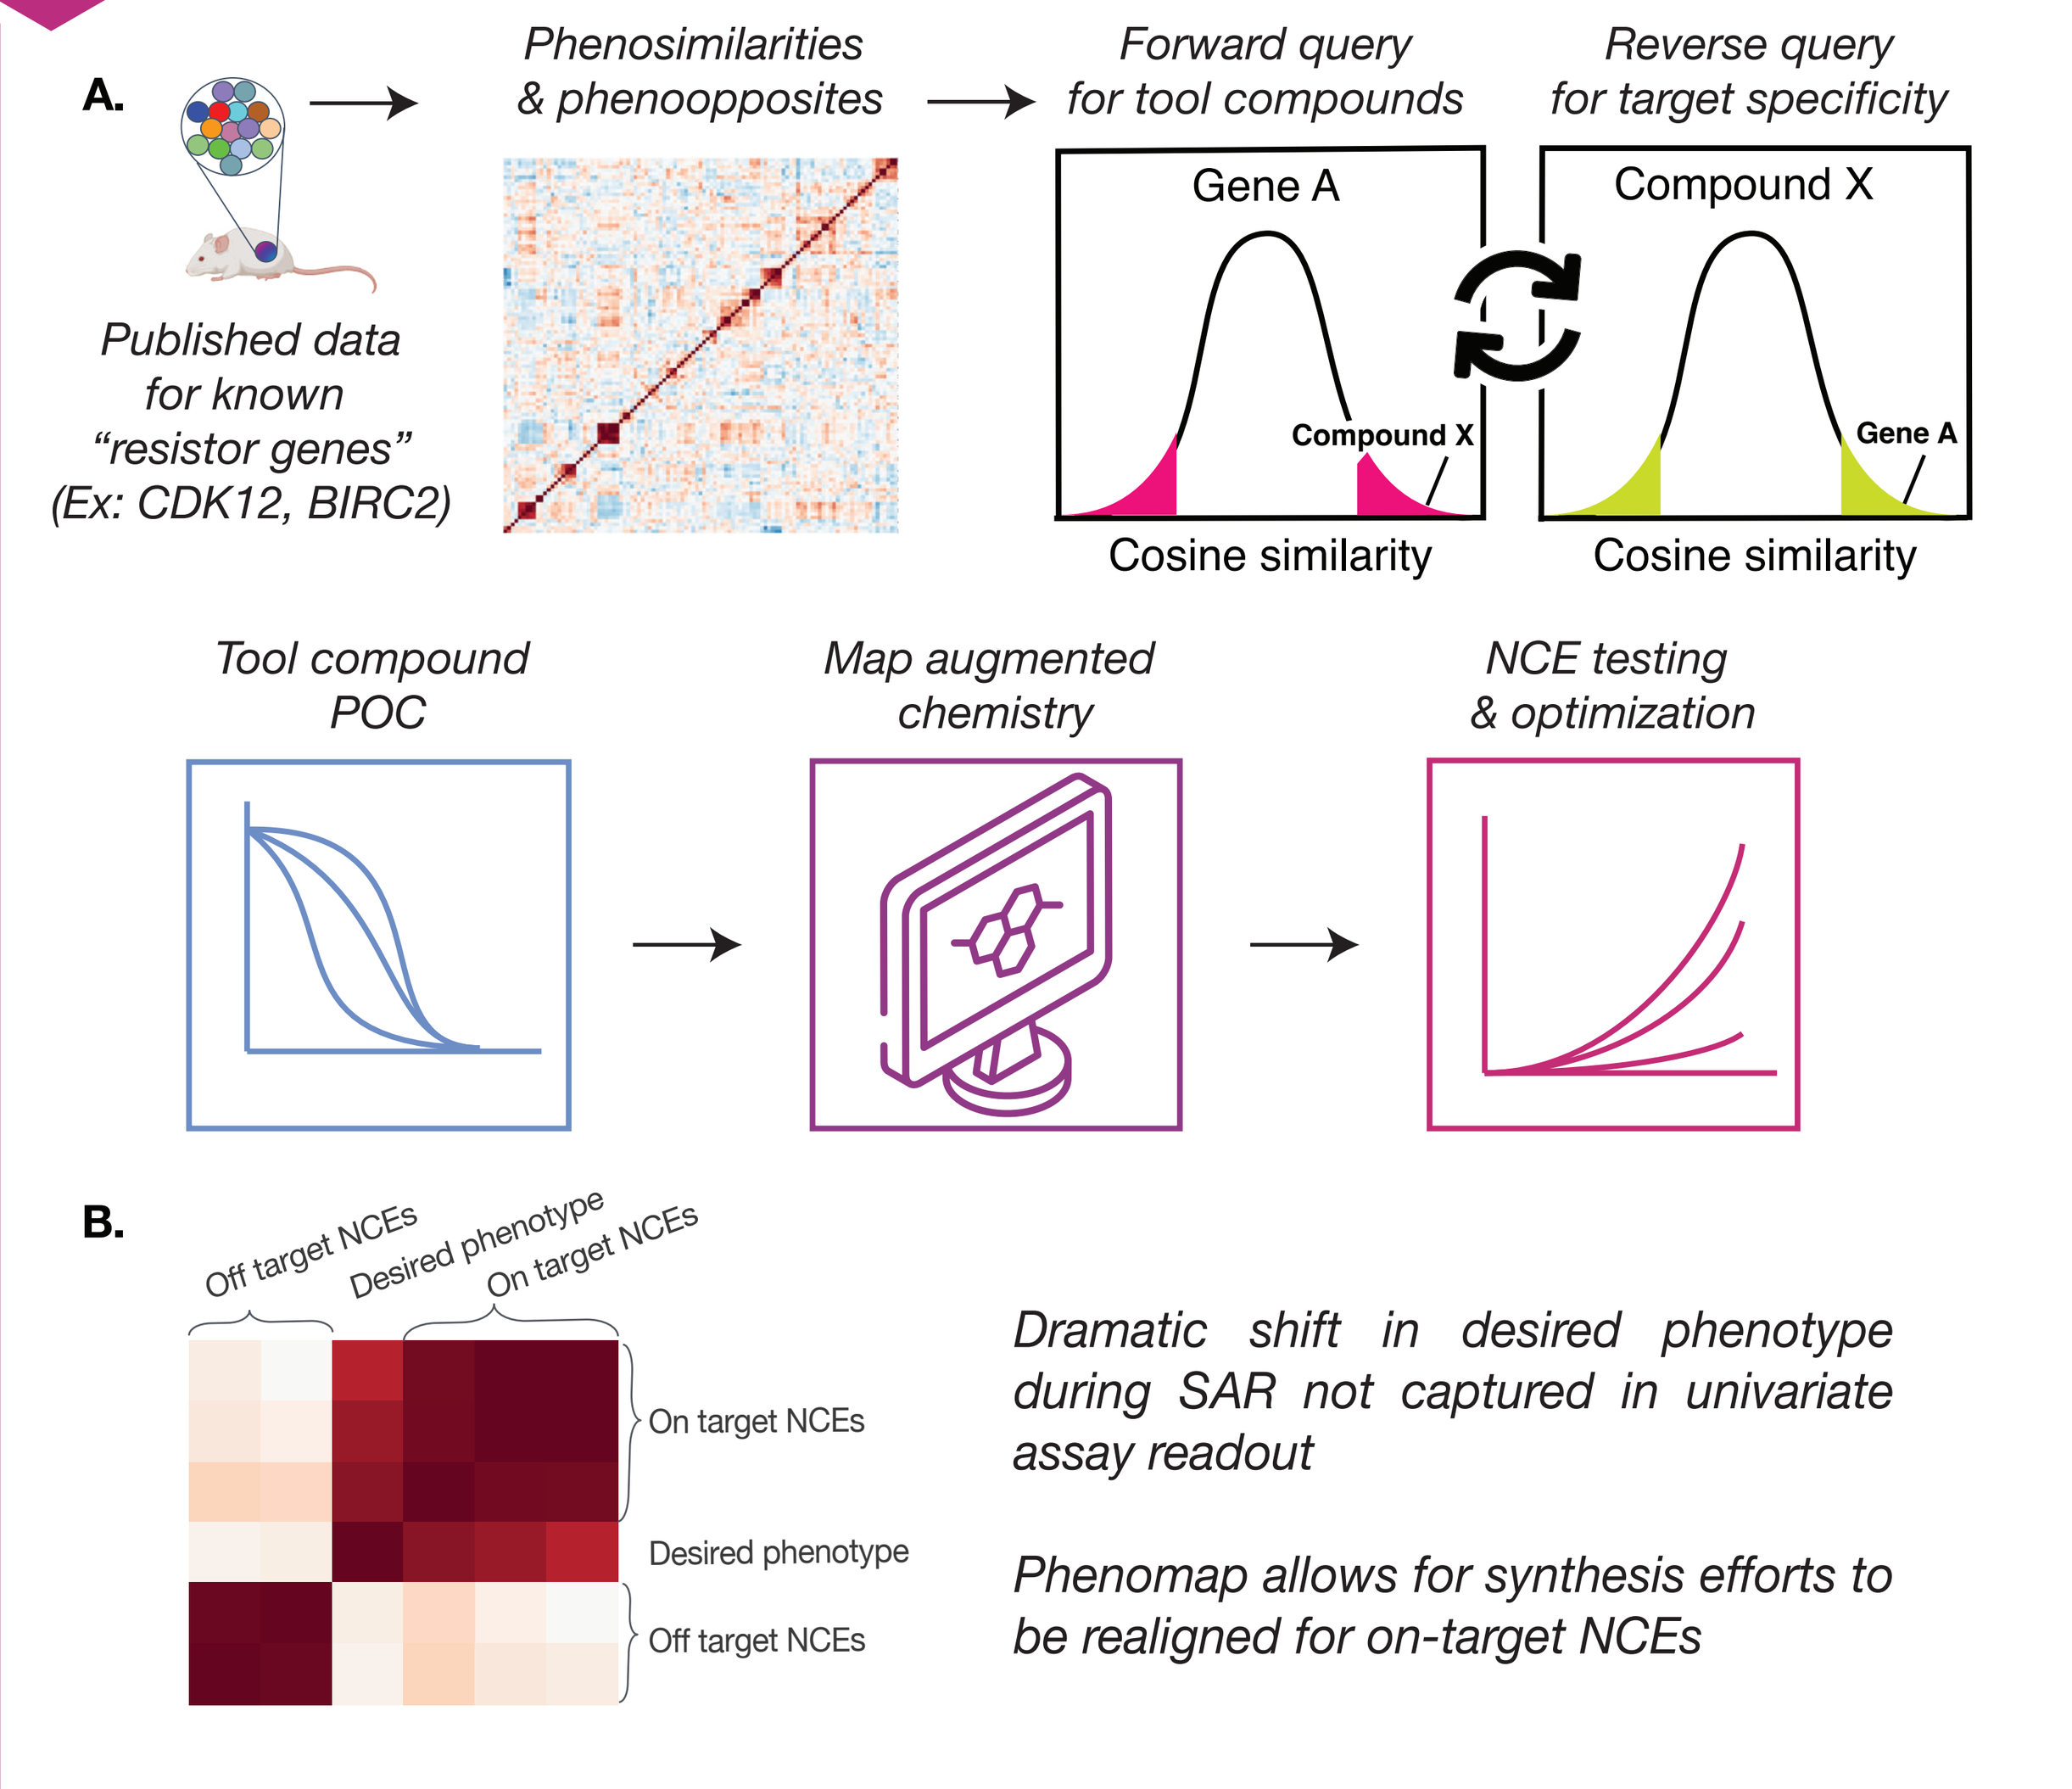

A.
Published data for known "resistor genes" (Ex: CDK12, BIRC2)
Phenosimilarities & phenoopposites
Forward query for tool compounds
Reverse query for target specificity
Gene A
Compound X
Compound X
Gene A
Cosine similarity
Cosine similarity
Tool compound POC
Map augmented chemistry
NCE testing & optimization
B.
Off target NCEs
Desired phenotype
On target NCEs
On target NCEs
Desired phenotype
Off target NCEs
Dramatic shift in desired phenotype during SAR not captured in univariate assay readout
Phenomap allows for synthesis efforts to be realigned for on-target NCEs

Figure 2: Using biological maps to identify novel targets and drive iterative chemical optimization. A, Workflow for the origin of two of Recursion's preclinical programs highlighted in Figures 3 and 4 below. We leverage published data sets for known tumor intrinsic "resistor genes" and query the Map for phenosimilars and pheno-opposites. Forward query refers to inputing the gene of interest to query for compounds that likely mimic or rescue the loss of that gene. Reverse query refers to inputing the compound of interest from the Forward query to determine how specific the compound is for that gene. It is through these features that we can discover novel uses for known chemical entities from which upon we can build novel chemistry through structure-activity relationships (SAR). B, Utilization of the map for driving SAR. NCEs are verified to sustain the desired phenotype, and NCEs that deviate phenomically are immediately de-prioritized.

3 NOVEL CHEMICAL ENTITY ENHANCES ANTI-PD1 RESPONSE AND DRIVES IMMUNOLOGICAL MEMORY WHILE ALSO REDUCING PERIPHERAL INFLAMMATION

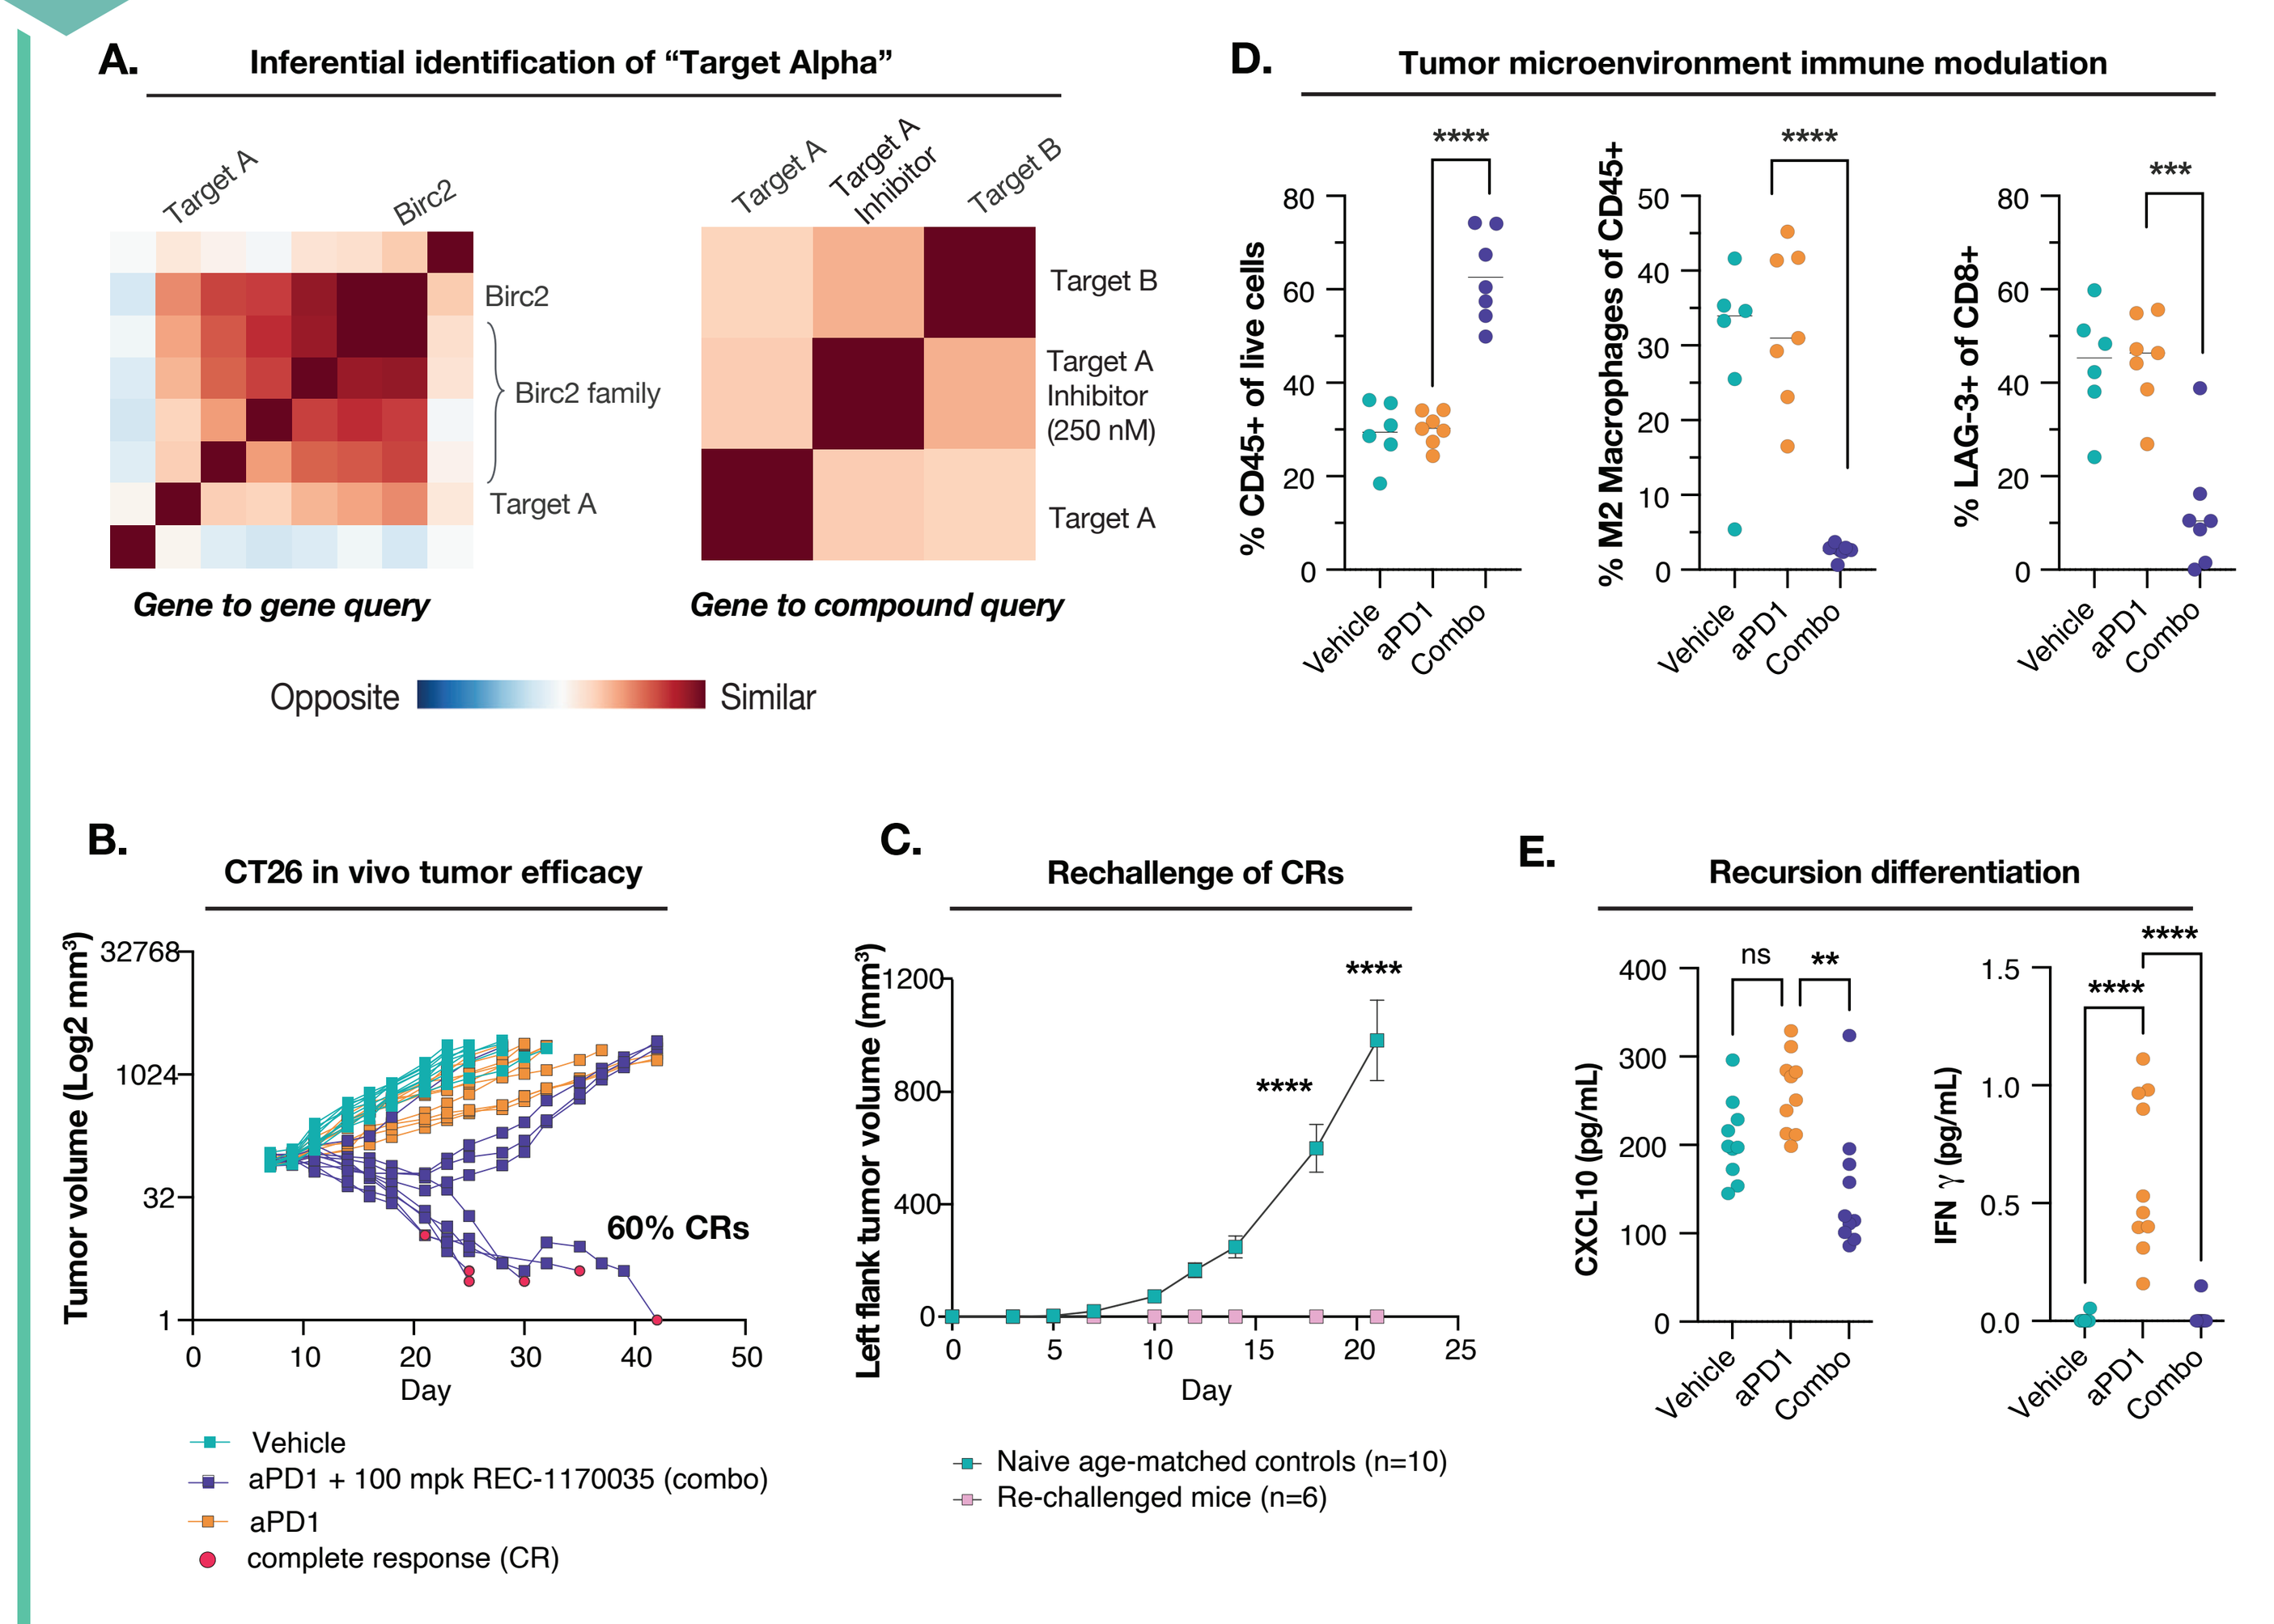

A.
Inferential identification of "Target Alpha"
Target A
Birc2
Birc2
Birc2 family
Target A
Target A
Target A Inhibitor
Target B
Target B
Target A Inhibitor (250 nM)
Target A
Gene to gene query
Gene to compound query
Opposite
Similar
D.
Tumor microenvironment immune modulation
% CD45+ of live cells
% M2 Macrophages of CD45+
% LAG-3+ of CD8+
Vehicle
aPD1
Combo
B.
CT26 in vivo tumor efficacy
Tumor volume (Log2 mm³)
Day
60% CRs
Vehicle
aPD1 + 100 mpk REC-1170035 (combo)
aPD1
complete response (CR)
C.
Rechallenge of CRs
Left flank tumor volume (mm³)
Day
Naive age-matched controls (n=10)
Re-challenged mice (n=6)
E.
Recursion differentiation
CXCL10 (pg/mL)
IFN γ (pg/mL)

Figure 3: Novel chemical entity enhances anti-PD1 response while suppressing peripheral inflammation. (A) Phenomap images demonstrating initial inference of phenosimilarity between Birc2 and Target (gene) A (left) and phenosimilarity between Target A Inhibitor ("the tool compound") and Targets A and B. Map augmented chemistry was performed to identify a new chemical entity (NCE) with better potency and efficacy over the tool compound (REC-1170035). (B) Mice bearing CT26 tumors treated with 100 mpk REC-1170035 (QD, PO) in combination with 10 mpk anti-PD1 (Q3D, IP) show 60% complete tumor regressions (CRs) compared to 10 mpk anti-PD1 treatment alone. (C) Mice from (A) harboring CRs were taken off drug for 30 days and then re-challenged with CT26 tumors in the opposite (left) flank and compared to naive, age-matched control animals. (D) Flow cytometric assessment of the CT26 tumor microenvironment after 11 days of treatment with vehicle,10 mpk anti-PD1, or 100 mpk REC-1170035 in combination with anti-PD1 ("combo"). (E) Compared to anti-PD1 alone, REC-1170035 reduces markers of peripheral inflammation (CXCL10, IFNg) while maintaining potent anti-tumor immunity. **p<0.01, ***p<0.001, ****p<0.0001.

4 TARGETING RBM39: A NOVEL CDK12-ADJACENT TARGET SENSITIZES HR-PROFICIENT OVARIAN CANCER TO PARP INHIBITION

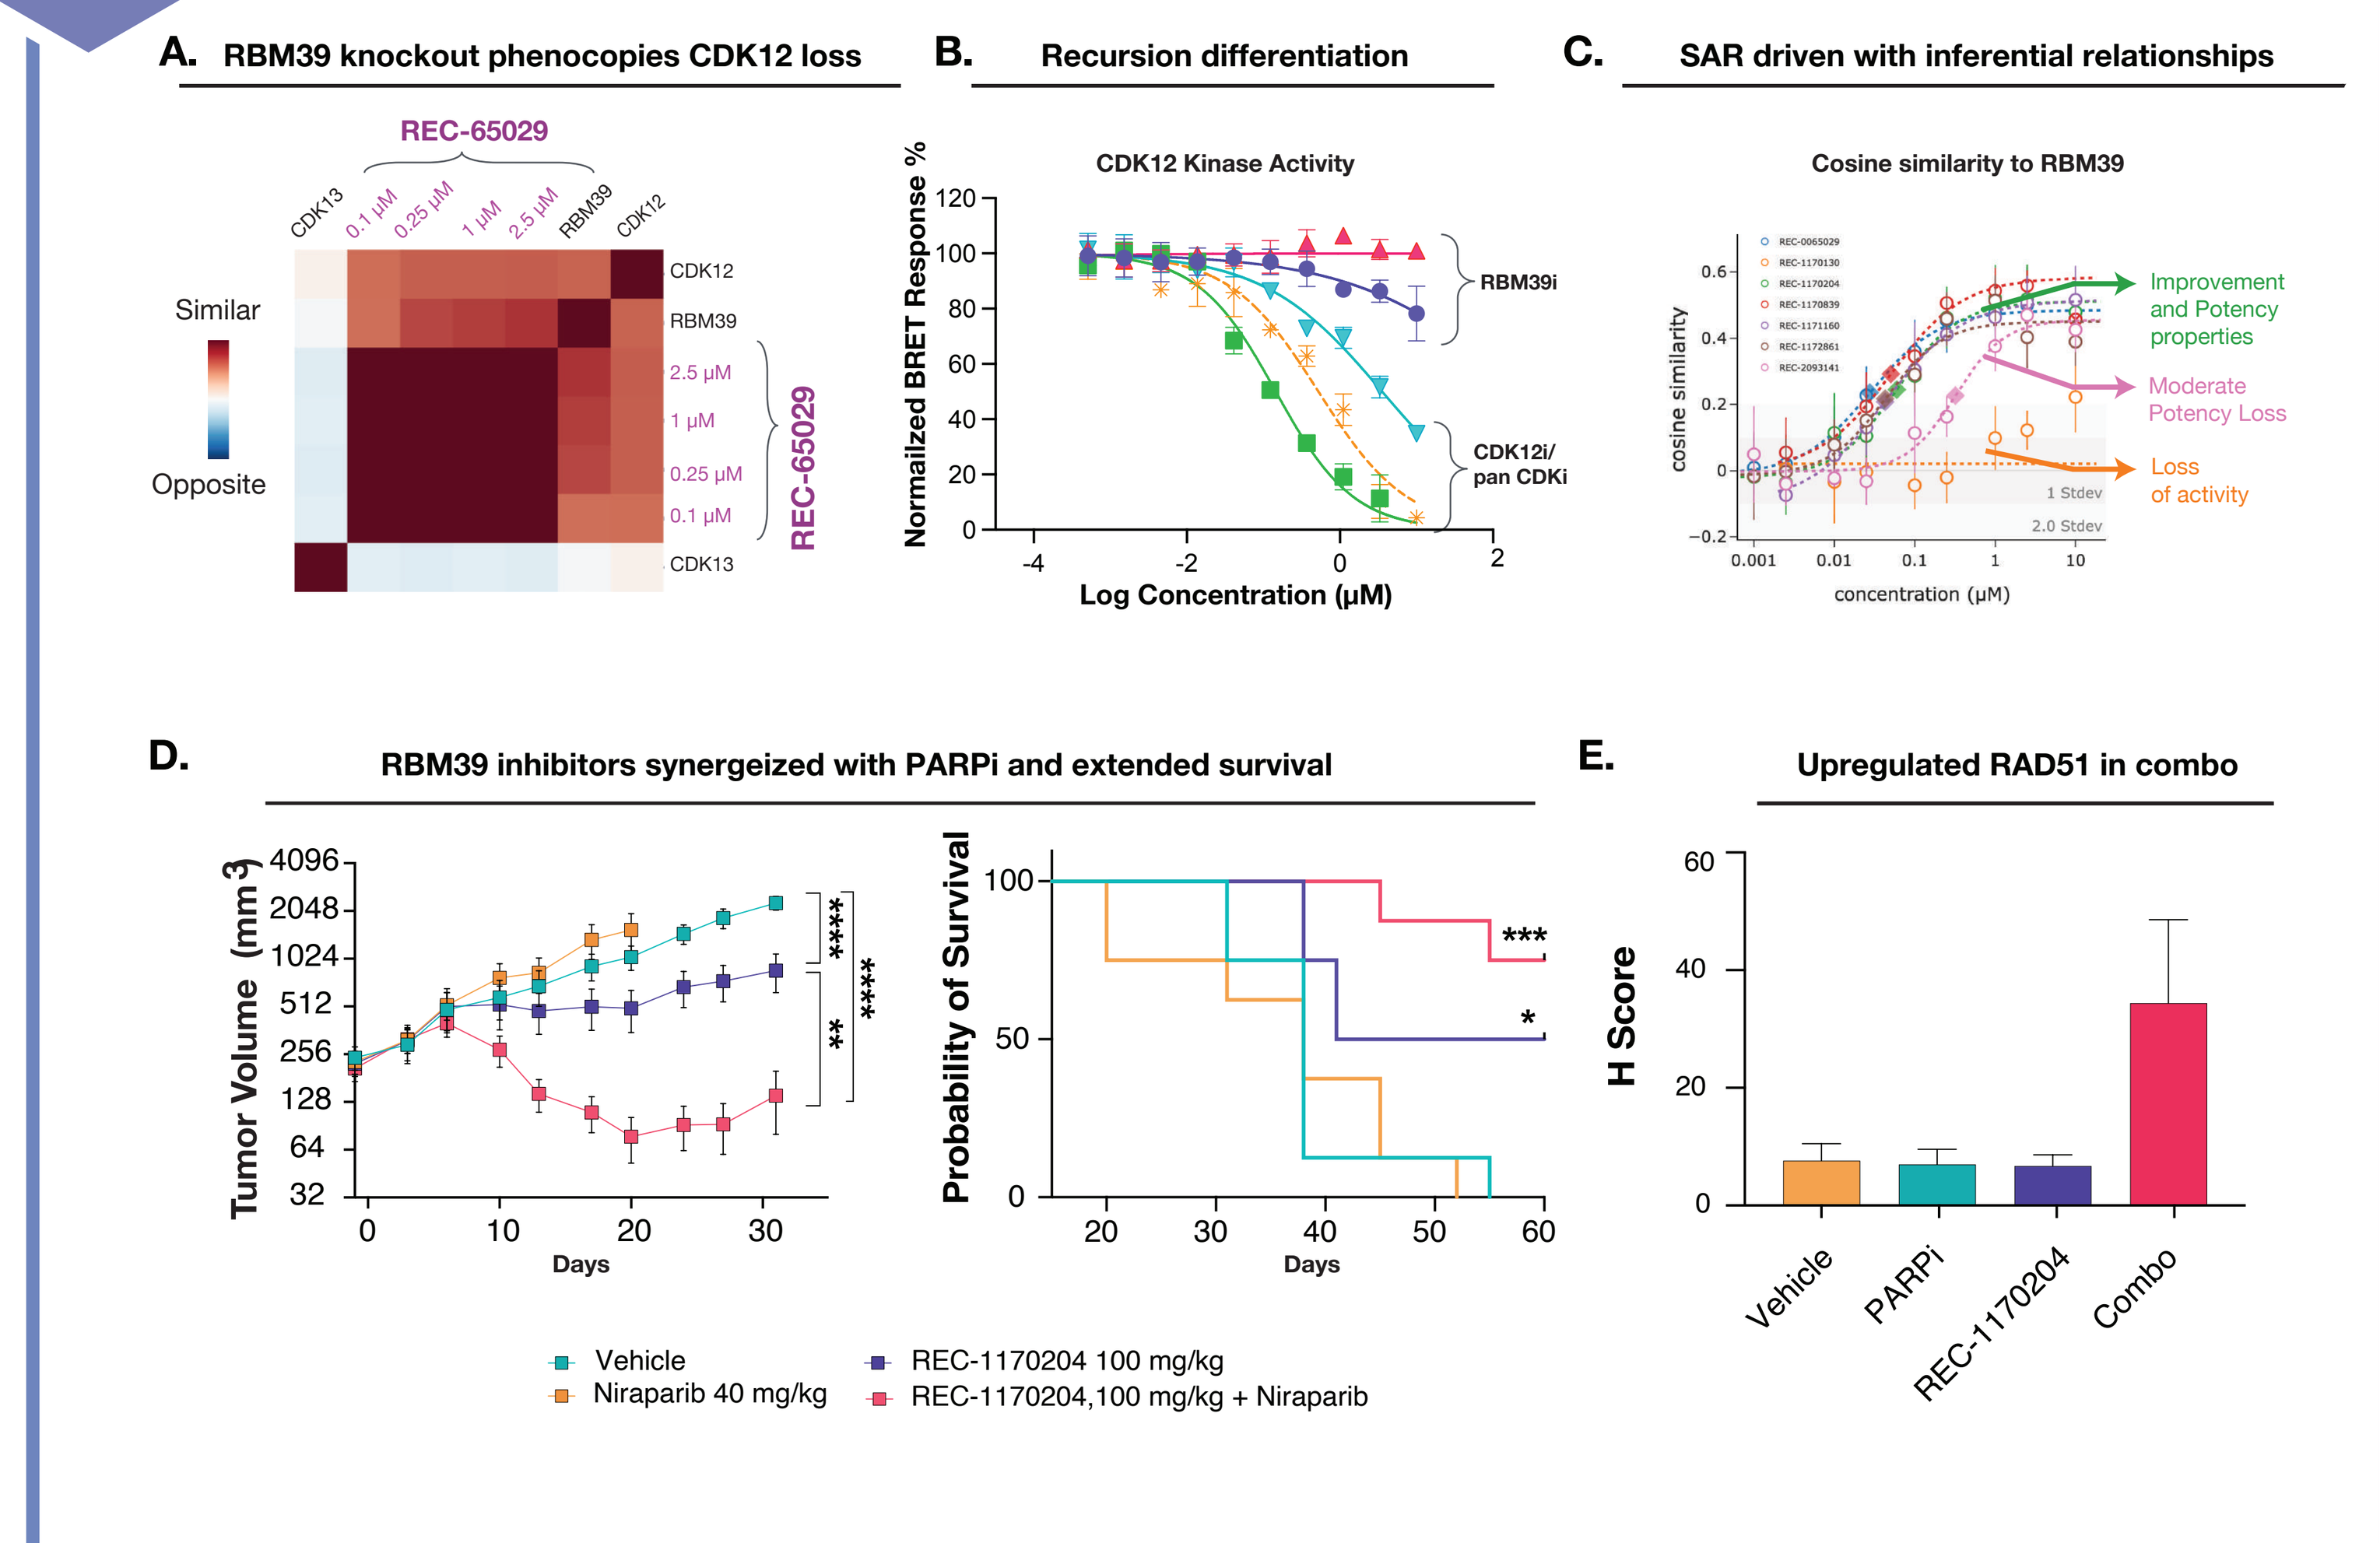

A. RBM39 knockout phenocopies CDK12 loss
REC-65029
Similar
Opposite
CDK12
RBM39
CDK13
B. Recursion differentiation
CDK12 Kinase Activity
Normalized BRET Response %
Log Concentration (µM)
RBM39i
CDK12i/ pan CDKi
C. SAR driven with inferential relationships
Cosine similarity to RBM39
cosine similarity
concentration (µM)
Improvement and Potency properties
Moderate Potency Loss
Loss of activity
D.
RBM39 inhibitors synergeized with PARPi and extended survival
Tumor Volume (mm³)
Days
Probability of Survival
Vehicle
Niraparib 40 mg/kg
REC-1170204 100 mg/kg
REC-1170204,100 mg/kg + Niraparib
E.
Upregulated RAD51 in combo
H Score
Vehicle
PARPi
REC-1170204
Combo

Figure 4: RBM39 targeted therapy potentiated PARP inhibitor in HR-proficient ovarian cancer OV0273 PDX model. (A) Novel Map insight demonstrating Phenotypic similarity between CDK12 loss and RBM39 knockout at multiple concentrations REC-65029 (tool compound) with little or no functional similarity to CDK13. (B) RBM39 inhibitors don't engage or impact CDK12 Kinase activity. (C) SAR profiling was performed on Recursion OS platform that helped accelerate lead identification.This approach enabled rapid progression from insight to in vivo efficacy with lead NCE in 6 months. (D) In the OV0273 PDX model mice were treated with representative Recursion lead compound REC-1170204 (100 mg/kg, BID, PO) +/- Niraparib (40 mg/kg, QD, PO) for 32 days. Single agent REC-1170204 in combination with Niraparib resulted in a statistically significant tumor growth inhibition versus either Niraparib or vehicle arms, also the combination treatment resulted in 4 CRs and significantly improved survival > 30 days post final dose *p values: < 0.05,**p < 0.01, ***p< 0.001, ****p< 0.0001. (E) PD data from tumors taken from Day-3, 4h post dosing (dosed equivalent to efficacy study in D) Tumor lysates were probed for RAD51, a DDR marker and RBM39. RAD51 expression was elevated in the combo arm that demonstrated significant synergy in efficacy study.

5 INDUSTRIALIZING DRUG DISCOVERY

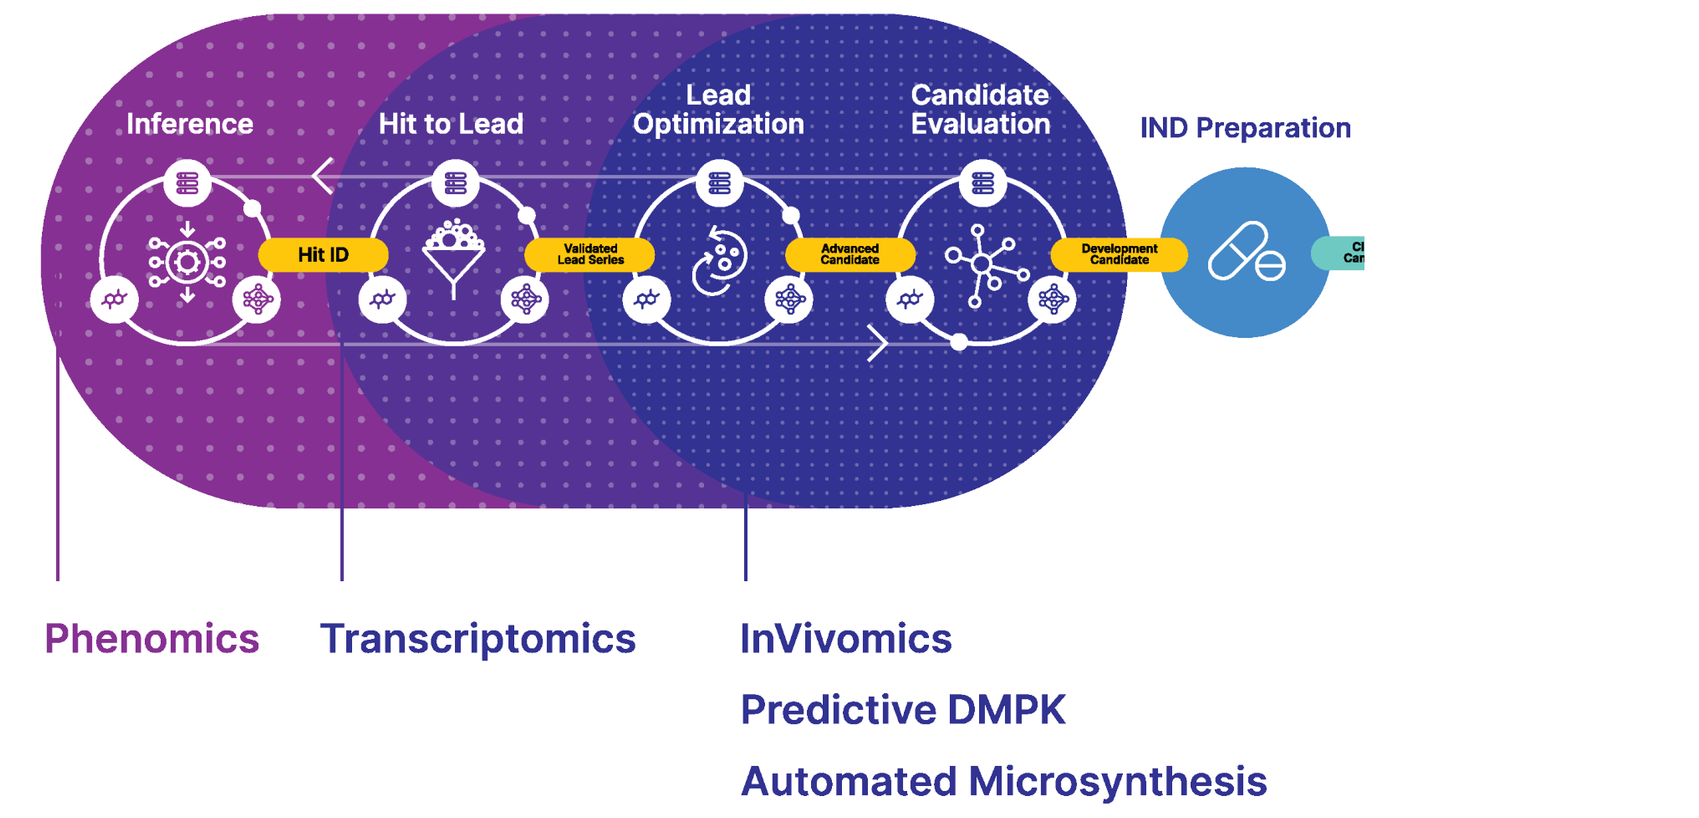

Inference
Hit to Lead
Lead Optimization
Candidate Evaluation
IND Preparation
Hit ID
Phenomics
Transcriptomics
InVivomics
Predictive DMPK
Automated Microsynthesis

Figure 5: Industrialized drug discovery. The industrialization and integration of various technological innovations across biology, chemistry, automation, data science, and engineering will ultimately modernize drug discovery and radically improve patient lives.

6 CONCLUSION

- Biological maps derived from high content imaging predict well appreciated and novel aspects of human biology. These maps enable us to identify gene-gene and gene-compound relationships. These inferences then translate to preclinical programs at a significantly faster cadence than traditional drug discovery approaches.
- Iterative structure activity relationship (SAR) cycling combined with map profiling allows us to drive chemical optimization from tool compounds to novel chemical entities (NCEs) with meaningful effect sizes in vivo.
- "Target alpha" was identified through query of the Recusion OS for immuno-oncology resistance genes. REC-1170035 induces 60% complete responses and immunological memory in combination with anti-PD1 while also reducing peripheral inflammatory cytokines, suggesting it may reduce the likelihood of immune related adverse events in the clinical setting.
- Recursion OS identified RBM39 as an alternative target to mimic CDK12 loss while mitigating pan CDK toxicities. RBM39 inhibitors induced multiple splicing alterations and downregulated DDR genes with unique MOA not linked to CDK12.
- The Recursion OS is meant to industrialize the drug discovery and development process through multiple cycles of learning and iteration.

7 ACKNOWLEDGEMENTS

Work presented here is made possible by the entire #OneRecursion team, from onboarding cells to map building to drug program development. Contributions span High Throughput Screening, Engineering, Data Science, Computational Biology, Discovery Biology, Medicinal Chemistry, PKPD, and many others. Special thanks to the OS Build anchor for helpful discussions and technical guidance.

8 REFERENCES

1. Cuccarese, *et al. bioRxiv.* 2020.
2. Recursion Pharmaceuticals, Inc. Form S-1 2021. Securities and Exchange Commission, 2021.
3. Manguso, *et al. Nature.* 2017.
4. Lawson, *et al. Nature.* 2020.
5. Recursion (2023) Download Day 2023 Full Day [Video] https://www.youtube.com/watch?v=NcxccxI8PWQ
6. Recursion (2023) Recursion's Mapping & Navigating Demonstration [Video] https://www.youtube.com/watch?v=tKYmBhUOP6k
7. Blucher, *et al.* Learning Meaningful Repesentations of Life Workshop, Conference on Neural Information Processing Systems. 2021.

Poster Location

Download day

Presented at the 2023 AACR Annual Meeting